ACP-204 Development Program for Alzheimer's Disease Psychosis: Population Pharmacokinetic Modeling

Mona Darwish,[1] Nancy Lin,[2] Bryan Dirks,[1] Amy Yamamoto,[3]* David Jaworowicz,[2] Kelly Maxwell,[2] Sanjeev Pathak[1]

[1]Acadia Pharmaceuticals Inc., Princeton, New Jersey; [2]Simulations Plus Inc., Research Triangle Park, North Carolina; [3]Acadia Pharmaceuticals Inc., San Diego, California

*Presenting author

930

INTRODUCTION

- Pharmacotherapy for Alzheimer's disease psychosis and Lewy Body Dementia psychosis traditionally involves off-label use of atypical antipsychotics despite limited evidence of efficacy and adverse effects such as cognitive or motor impairment[1]
- Pimavanserin, a selective 5-HT_{2A} receptor inverse agonist/antagonist, is the only US Food and Drug Administration–approved treatment for hallucinations and delusions associated with Parkinson's disease[2,3]
- ACP-204, a similar 5-HT_{2A} receptor inverse agonist, is under investigation for treatment of Alzheimer's disease psychosis
- The current study aimed to develop and refine a population pharmacokinetics (popPK) model for ACP-204, identify intrinsic and extrinsic factors affecting PK variability, and assess the clinical relevance of these effects in a large virtual population

METHODS

- A popPK model was developed (**Figure 1**), and the clinical relevance of intrinsic and extrinsic factors on steady-state exposures (maximum plasma concentration at steady state [$C_{max,ss}$] and area under the plasma concentration-time curve over the last 24-hour dosing interval at steady state [$AUC_{0\text{-}24,ss}$]) was evaluated using a virtual population (n=1000; 60 mg/d) based on geometric mean ratios and 90% CI within a 0.8-1.25 boundary[4]

Figure 1. Development of a PopPK Model for ACP-204 in Adults

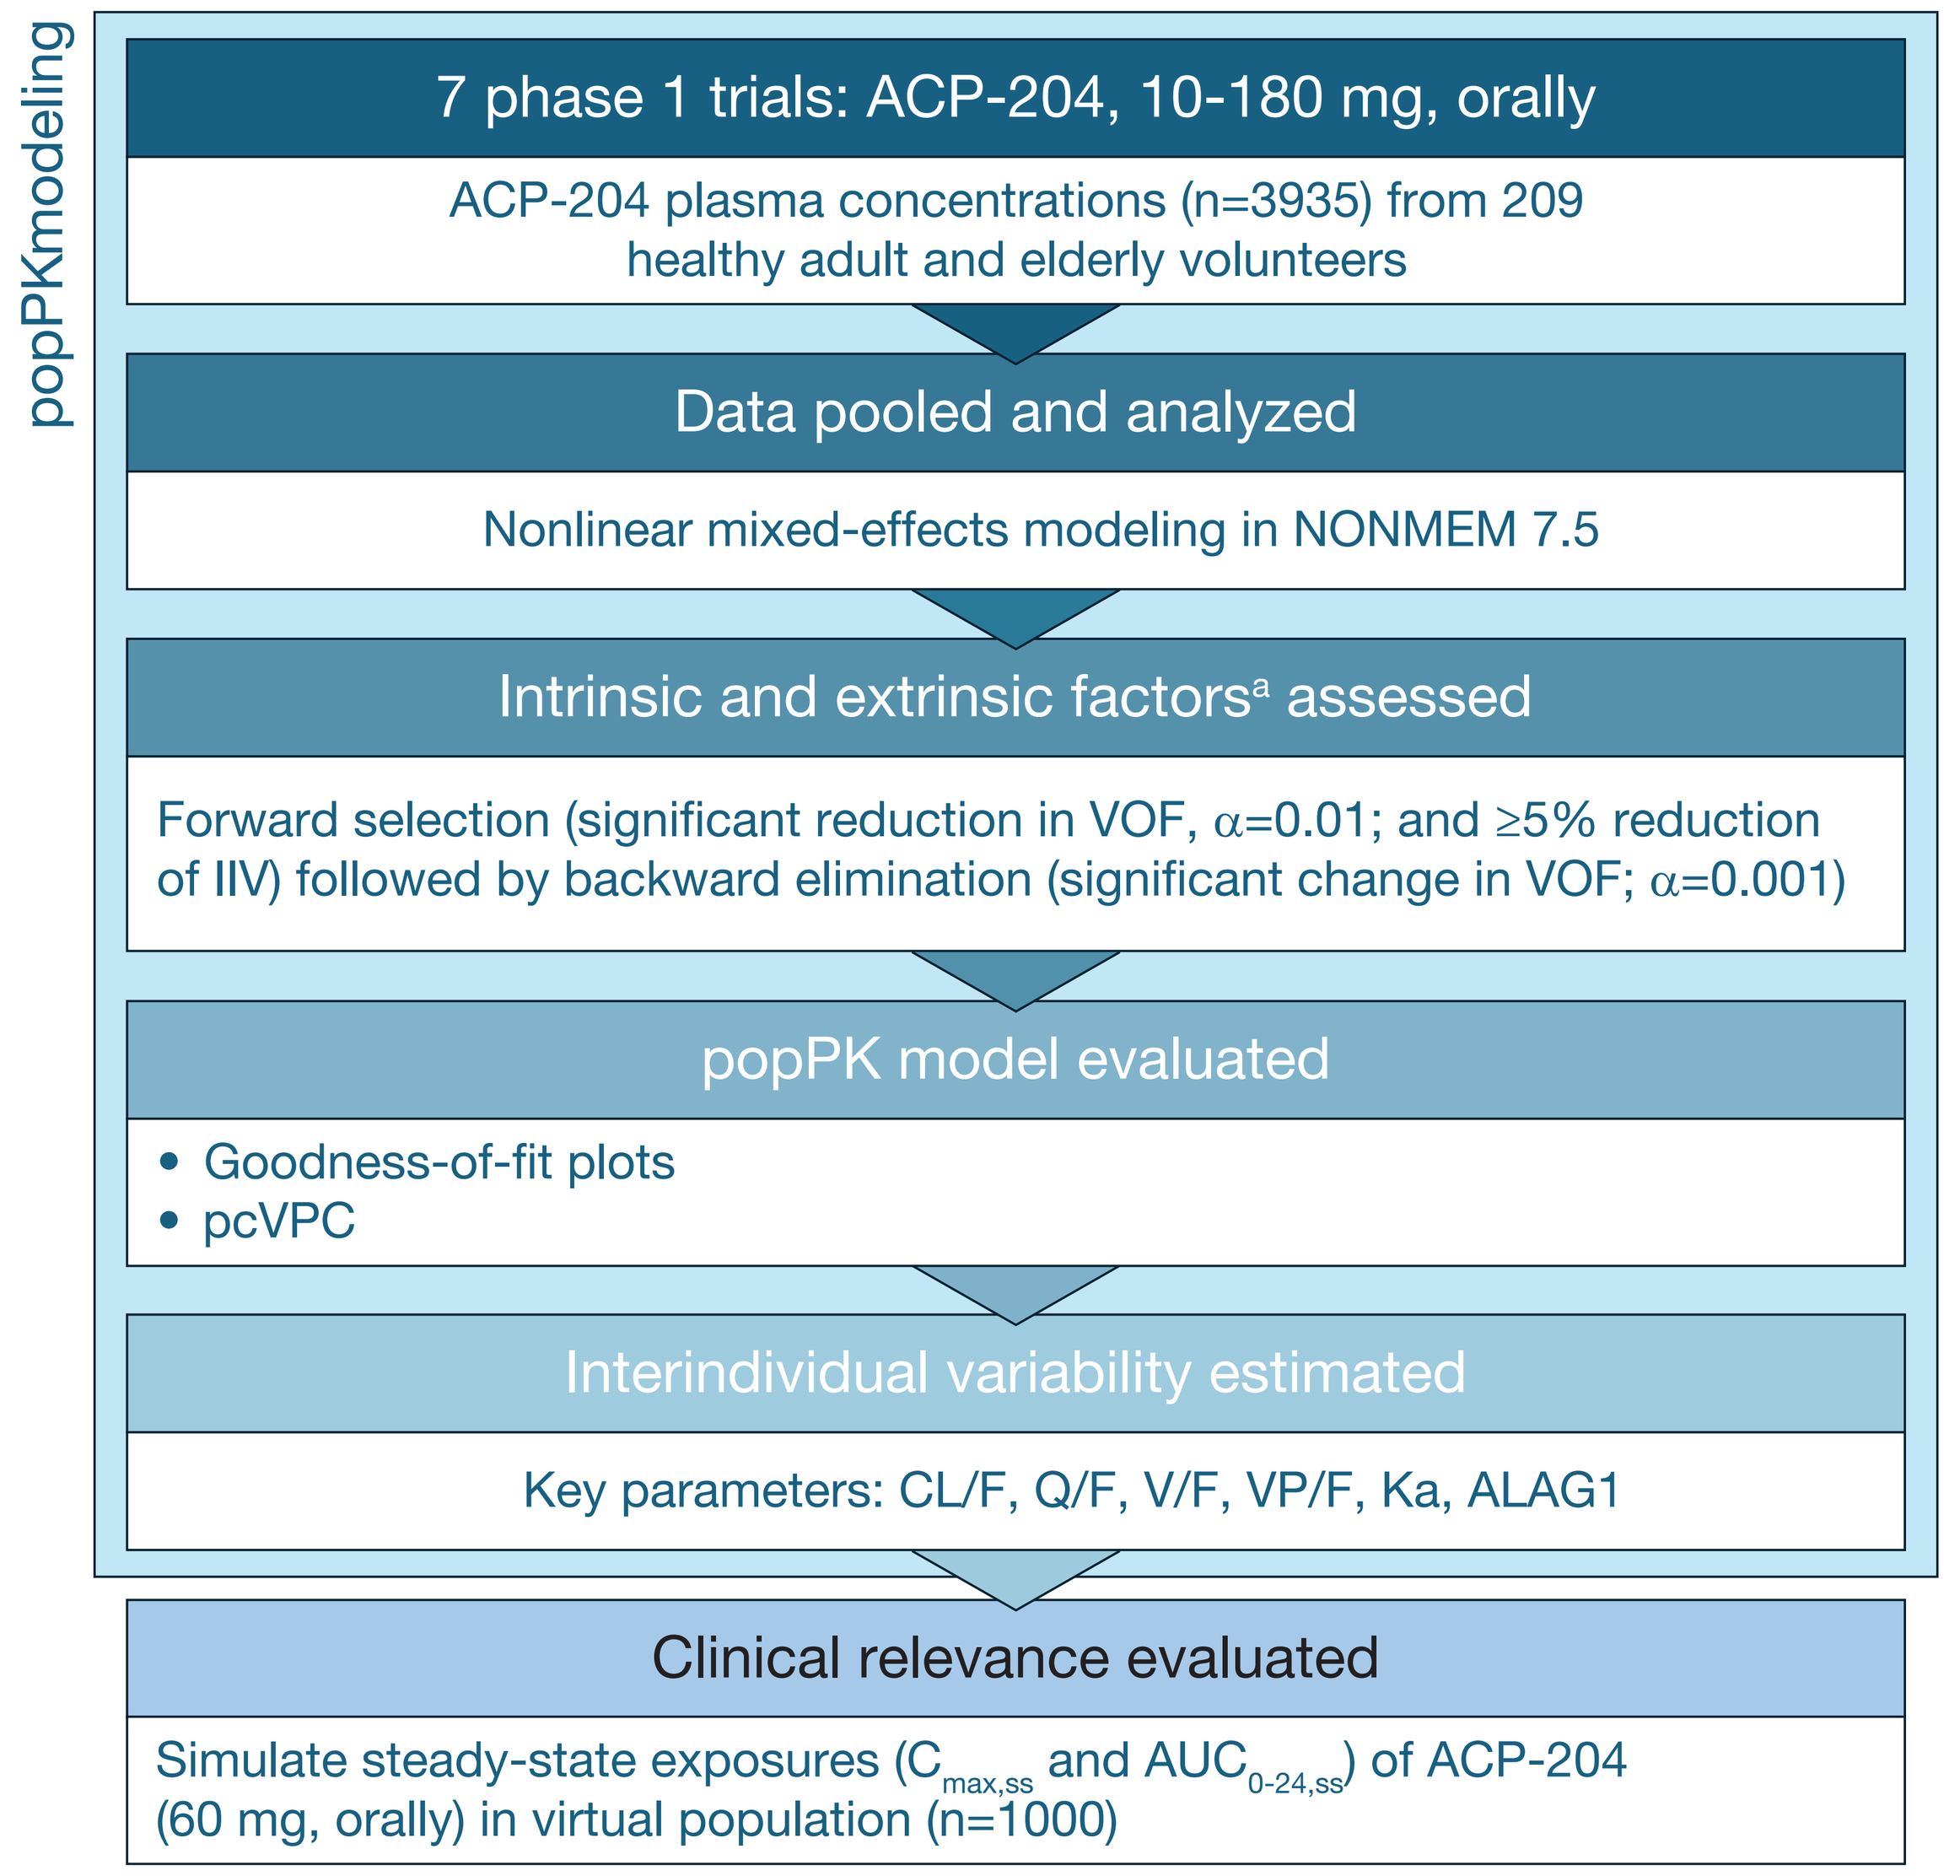
popPKmodeling
7 phase 1 trials: ACP-204, 10-180 mg, orally
ACP-204 plasma concentrations (n=3935) from 209 healthy adult and elderly volunteers
Data pooled and analyzed
Nonlinear mixed-effects modeling in NONMEM 7.5
Intrinsic and extrinsic factors[a] assessed
Forward selection (significant reduction in VOF, α=0.01; and ≥5% reduction of IIV) followed by backward elimination (significant change in VOF; α=0.001)
popPK model evaluated
• Goodness-of-fit plots
• pcVPC
Interindividual variability estimated
Key parameters: CL/F, Q/F, V/F, VP/F, Ka, ALAG1
Clinical relevance evaluated
Simulate steady-state exposures ($C_{max,ss}$ and $AUC_{0\text{-}24,ss}$) of ACP-204 (60 mg, orally) in virtual population (n=1000)

ALAG1, lag time for first-order absorption; $AUC_{0\text{-}24,ss}$, area under the plasma concentration-time curve over the last 24-hour dosing interval at steady state; BMI, body mass index; CL/F, apparent central clearance; $C_{max,ss}$, maximum plasmaconcentration at steady state; IIV, interindividual variability; Ka, absorption rate constant; pcVPC, prediction-corrected visual predictive check; PK, pharmacokinetic; popPK, population pharmacokinetics; Q/F, apparent intercompartment clearance; V/F, apparent central volume of distribution; VOF, value of the objective function; VP/F, apparent peripheral volume of distribution.
[a]Via covariate analysis; factors included age, weight, BMI, sex, race, liver and kidney function, and fed/fasted status.

RESULTS

- The final popPK model for ACP-204 was a 1-compartment model with first-order absorption with lag time and linear elimination
- Categorical factors of food status (fed vs fasted) and sex (male vs female) were statistically significant (*P*<0.001) predictors of ACP-204 PK
 - Each explained ≥5% of interindividual variability in the parameter of interest
 - However, there was no clinically relevant impact on $C_{max,ss}$ or $AUC_{0\text{-}24,ss}$
- Prediction-corrected visual predictive checks confirmed that the model adequately captured central tendency and variability in ACP-204 concentrations in the virtual population (**Figure 2**)

Figure 2. Prediction-Corrected Visual Predictive Check Plots for the Final PopPK Model, Stratified by Fasted (A) vs Fed (B) Status and Male (C) vs Female (D) Sex

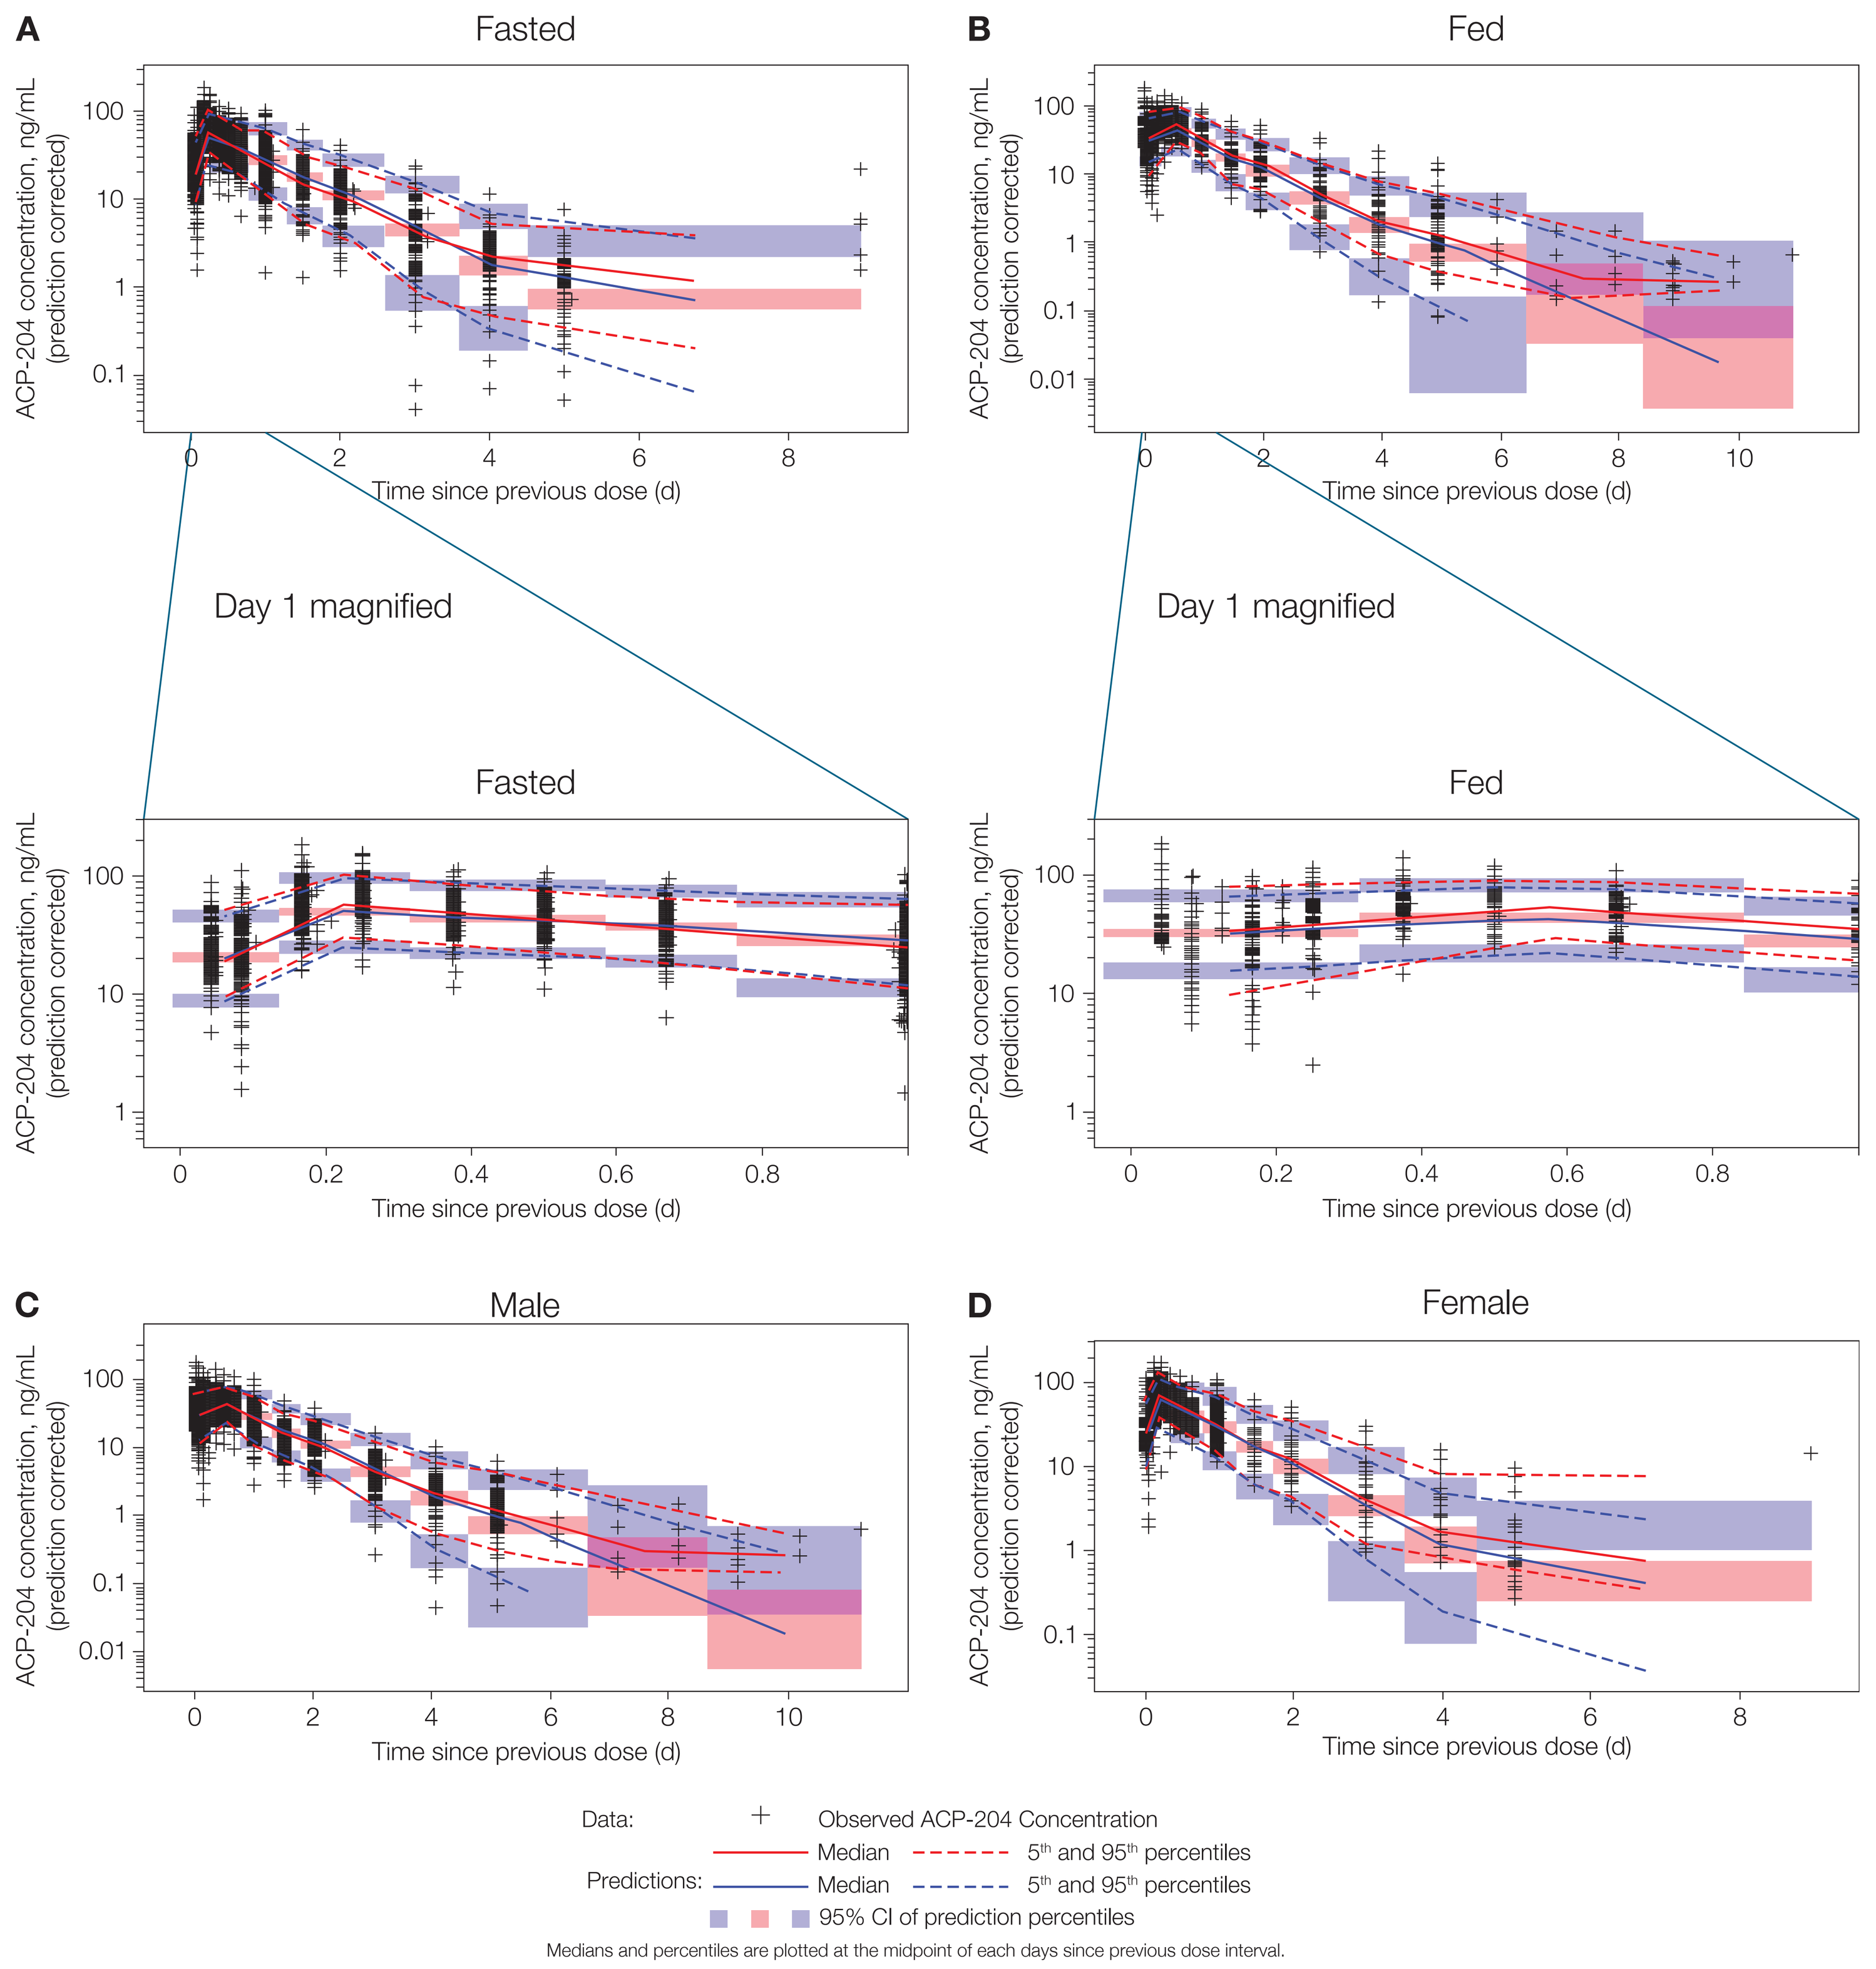
A
Fasted
B
Fed
Day 1 magnified
Fasted
Fed
C
Male
D
Female
ACP-204 concentration, ng/mL (prediction corrected)
Time since previous dose (d)
Data: Observed ACP-204 Concentration
Predictions: Median 5th and 95th percentiles
Median 5th and 95th percentiles
95% CI of prediction percentiles
Medians and percentiles are plotted at the midpoint of each days since previous dose interval.

PopPK, population pharmacokinetics.

- The forest plot assessment of covariates on $C_{max,ss}$ and $AUC_{0\text{-}24,ss}$ suggests that both fed/fasted status and sex do not exhibit a clinically relevant effect on steady-state ACP-204 exposures (**Figure 3**)

Figure 3. Clinical Relevance of Covariate Effects of Sex and Fed Status on $C_{max,ss}$ (A) and $AUC_{0\text{-}24,ss}$ (B) in the Final PopPK Model[a,b]

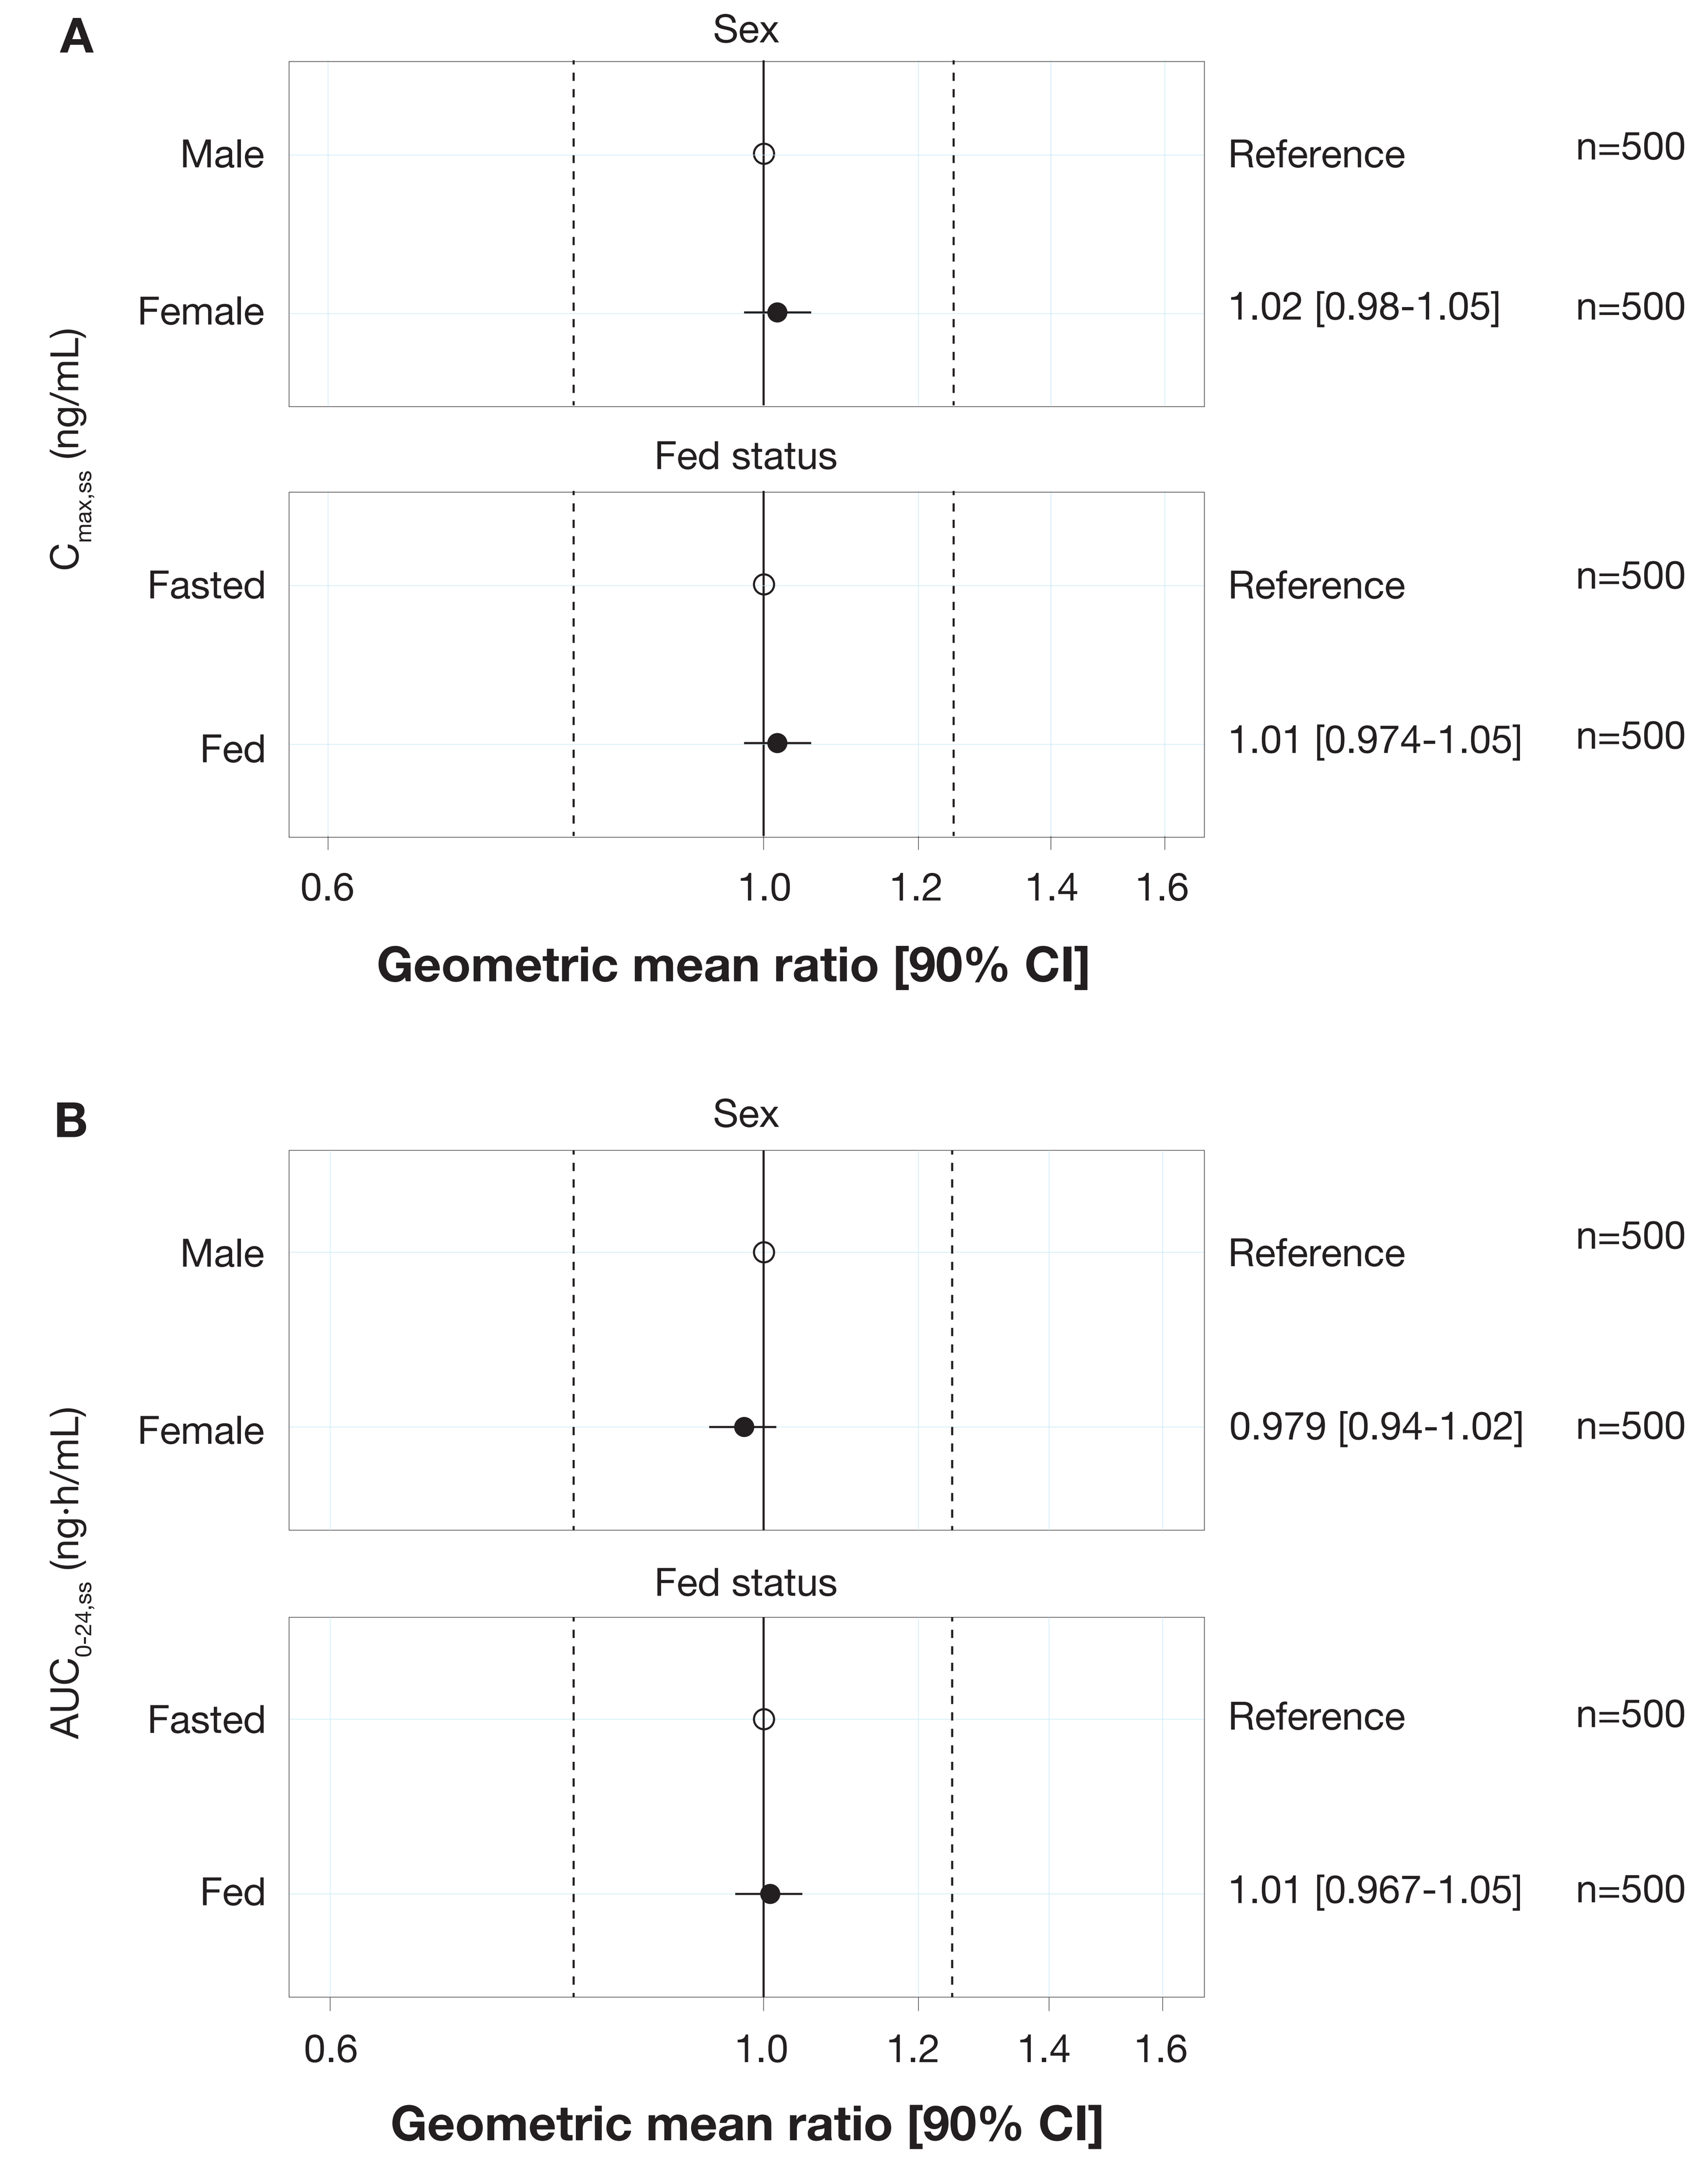
A
$C_{max,ss}$ (ng/mL)
Sex
Male Reference n=500
Female 1.02 [0.98-1.05] n=500
Fed status
Fasted Reference n=500
Fed 1.01 [0.974-1.05] n=500
0.6 1.0 1.2 1.4 1.6
Geometric mean ratio [90% CI]
B
$AUC_{0\text{-}24,ss}$ (ng·h/mL)
Sex
Male Reference n=500
Female 0.979 [0.94-1.02] n=500
Fed status
Fasted Reference n=500
Fed 1.01 [0.967-1.05] n=500
0.6 1.0 1.2 1.4 1.6
Geometric mean ratio [90% CI]

$AUC_{0\text{-}24,ss}$, area under the plasma concentration-time curve over the last 24-hour dosing interval at steady state; $C_{max,ss}$, maximum plasma concentration at steady state; popPK, population pharmacokinetics.
[a]Brackets indicate the respective limit is included in the interval; parentheses indicate the respective limit is not included in the interval.
[b]Vertical dashed lines at 0.8 and 1.25 are clinical relevance bounds.

- The estimated population mean absorption rate constant (Ka) of 0.692/h describes a first-order absorption half-life of approximately 1 hour for a typical participant (**Table 1**)

Table 1. Parameter Estimates and Standard Errors of the Final ACP-204 popPK Model

Parameter		Final parameter estimate: Population mean (% RSE)	Magnitude of variability: Final estimate, % CV (% RSE)
CL/F	Apparent oral clearance, L/d	849 (2.84)	42.6[a] (10.3)
V/F	Apparent central volume of distribution, L	930 (2.43)	32.1[b] (11.3)
	Additive shift in V/F for female participants	–131 (16.8)	
Ka	First-order absorption rate constant, 1/d	16.6 (4.65)	67.5 (15.2)
ALAG1	Absorption lag time, d	0.0300 (1.91)	10.7 (22.0)
	Proportional shift in ALAG1 for fed status	0.290 (10.4)	
Covariance (IIV in V/F, IIV in CL/F)		0.109[a,b,c] (11.9)	
Residual variability	Proportional	0.0388 (6.38)	19.7
Minimum VOF=15,834.209			

ALAG1, absorption lag time for a first-order absorption process; CL/F, apparent oral clearance; CV, coefficient of variation; IIV, interindividual variability; Ka, absorption rate constant; NA, not applicable; popPK, population pharmacokinetics; RSE, relative standard error; V/F, apparent volume of distribution after extravascular administration; VOF, value of the objective function.
[a]The following parameter estimates were found to be highly correlated (r^2≥0.81): covariance (IIV in V/F, IIV in CL/F), IIV in CL/F.
[b]The following parameter estimates were found to be highly correlated (r^2≥0.81): IIV in V/F, covariance (IIV in V/F, IIV in CL/F).
[c]The calculated correlation coefficient (r) associated with covariance (IIV in V/F, IIV in CL/F) was 0.852 with r^2=0.726.

LIMITATIONS

- Phase 1 trials in healthy participants (predominantly White and male; median age of 37 years) may affect the generalizability of results

CONCLUSIONS

- Following oral administration from 10 to 180 mg, the PK of ACP-204 is adequately described by a 1-compartment model having first-order absorption with lag time and linear elimination kinetics
- Intrinsic and extrinsic factors assessed—including age, body mass index, race, sex, liver and kidney function, and fasted/fed status—were not found to have a clinically relevant effect on steady-state ACP-204 exposures

Acknowledgments

Medical writing support was provided by Phillip Lewis, PhD, from Citrus Health Group, Inc. (Chicago, Illinois), and was funded by Acadia Pharmaceuticals, Inc., San Diego, California.

Author Disclosures

MD, **BD**, **AY**, and **SP** are employees of Acadia Pharmaceuticals, Inc., and may own stock and/or stock options. **NL**, **DJ**, and **KM** are employees of Simulations Plus, Inc., and may own stock and/or stock options.

References

1. Tariot PN, et al. *N Engl J Med.* 2021;385(4):309-319.
2. Acadia Pharmaceuticals Inc. NUPLAZID® (pimavanserin) Prescribing Information. 2025. Accessed September 9, 2025, https://www.nuplazid.com/pdf/nuplazid-prescribing-information.pdf
3. Berrio A, et al. *Clin Park Relat Disord.* 2025;12:100342.
4. Menon-Andersen D, et al. *Clin Pharmacol Ther.* 2011;90(3):471-474.

Presented at the 2025 American College of Clinical Pharmacy (ACCP) Annual Meeting; October 18-21, 2025; Minneapolis, MN

ACADIA